2593

Association of an Inflammatory Gene Signature With CD8 Expression by Immunohistochemistry in Multiple Tumor Types

Peter M. Szabo,[1] Zhenhao Qi,[1,*] Scott Ely,[1] Robin Edwards,[1] James Lu,[2] James Cooley,[2] Marian Navratil,[2] Gillian L. Dalgliesh,[3] Neeraj Adya[1]

[1]Bristol-Myers Squibb, Princeton, NJ, USA; [2]HTG Molecular Diagnostics Inc, Tucson, AZ, USA; [3]QIAGEN Manchester Ltd, Manchester, UK

*At the time the analysis was conducted

Background

- An inflammatory phenotype in the tumor microenvironment (TME) has been associated with improved clinical outcomes in patients treated with immuno-oncology (I-O) therapy across multiple tumor types[1]
 - Infiltration of CD8+ T cells is one of the surrogate markers for inflammation and can be assessed by employing immunohistochemistry (IHC) assays.[2,3] However, IHC assays have limited capability to simultaneously interrogate multiple biomarkers
- Analyzing the transcriptome by gene expression profiling (GEP) can be used to identify signatures predictive of response to I-O therapy in patients with cancer[4,5]
 - A multiparameter tumor inflammation assay based on GEP may offer a more robust characterization of inflammation by simultaneously interrogating multiple genes, thereby extending the utility of single-gene expression analysis or protein-based IHC assessment to characterize the TME

Objectives

- Develop a GEP-based, investigational-use-only (IUO) assay to interrogate inflammation in the TME
- Evaluate and analytically validate a gene expression signature for inflammation that correlates with CD8 expression by IHC via pathology-assisted digital scoring (CD8 IHC)
- Assess the prevalence of inflammation characterized by CD8+ T cells in the TME across multiple tumor types, using IHC and GEP with the derived CD8 signature

Methods

- GEP was performed on a set of tumor sections by next-generation sequencing (NGS) using an inflammation gene panel (inflammation panel)
 - The inflammation panel comprises 95 genes, including genes related to tumor inflammation and other I-O genes of interest, housekeeping genes, and control genes. This inflammation panel measures mRNA expression levels of all 95 genes on the panel
 - NGS data were filtered based on quality control metrics examined in the following order: input RNA quantity/quality, sequencing read depth, and samples with minimum expression variability
 - Of 1778 commercially procured formalin-fixed, paraffin-embedded (FFPE) samples, 1633 (91.84%) passed quality control and were used for inflammation analyses
- Using a machine-learning approach, the combination of genes that gave the best correlation with CD8 IHC was first developed for melanoma (MEL; n = 335) and squamous cell carcinoma of the head and neck (SCCHN; n = 392) independently and then using pooled MEL and SCCHN samples
 - Based on the 2 tumor-specific signatures and the signature derived from the pooled samples, a 16-gene CD8 signature was selected and subsequently verified across 10 additional tumor types (**Figure 1**)
- Each gene in the CD8 signature was assigned a specific weight on the inflammation panel
 - The output was a gene signature score for each sample tested across 12 tumor types representing a range of inflammation values
- Accuracy of the CD8 signature to predict CD8 IHC was assessed across 12 tumor types
 - CD8 IHC was performed by Mosaic Laboratories (Lake Forest, CA) using a monoclonal CD8 (clone C8/144B) antibody (Dako, an Agilent Technologies Co, Santa Clara, CA)
 - The 95% confidence interval (CI) for prediction of CD8 IHC was computed from the multiple linear regression model obtained using MEL and SCCHN samples
 - The model was considered accurate if ~ 95% of samples were contained within this CI
 - The relationship between CD8 IHC and the CD8 signature score was investigated by Pearson's correlation for each tumor type

Figure 1. Developing a multitumor CD8 inflammatory signature based on GEP

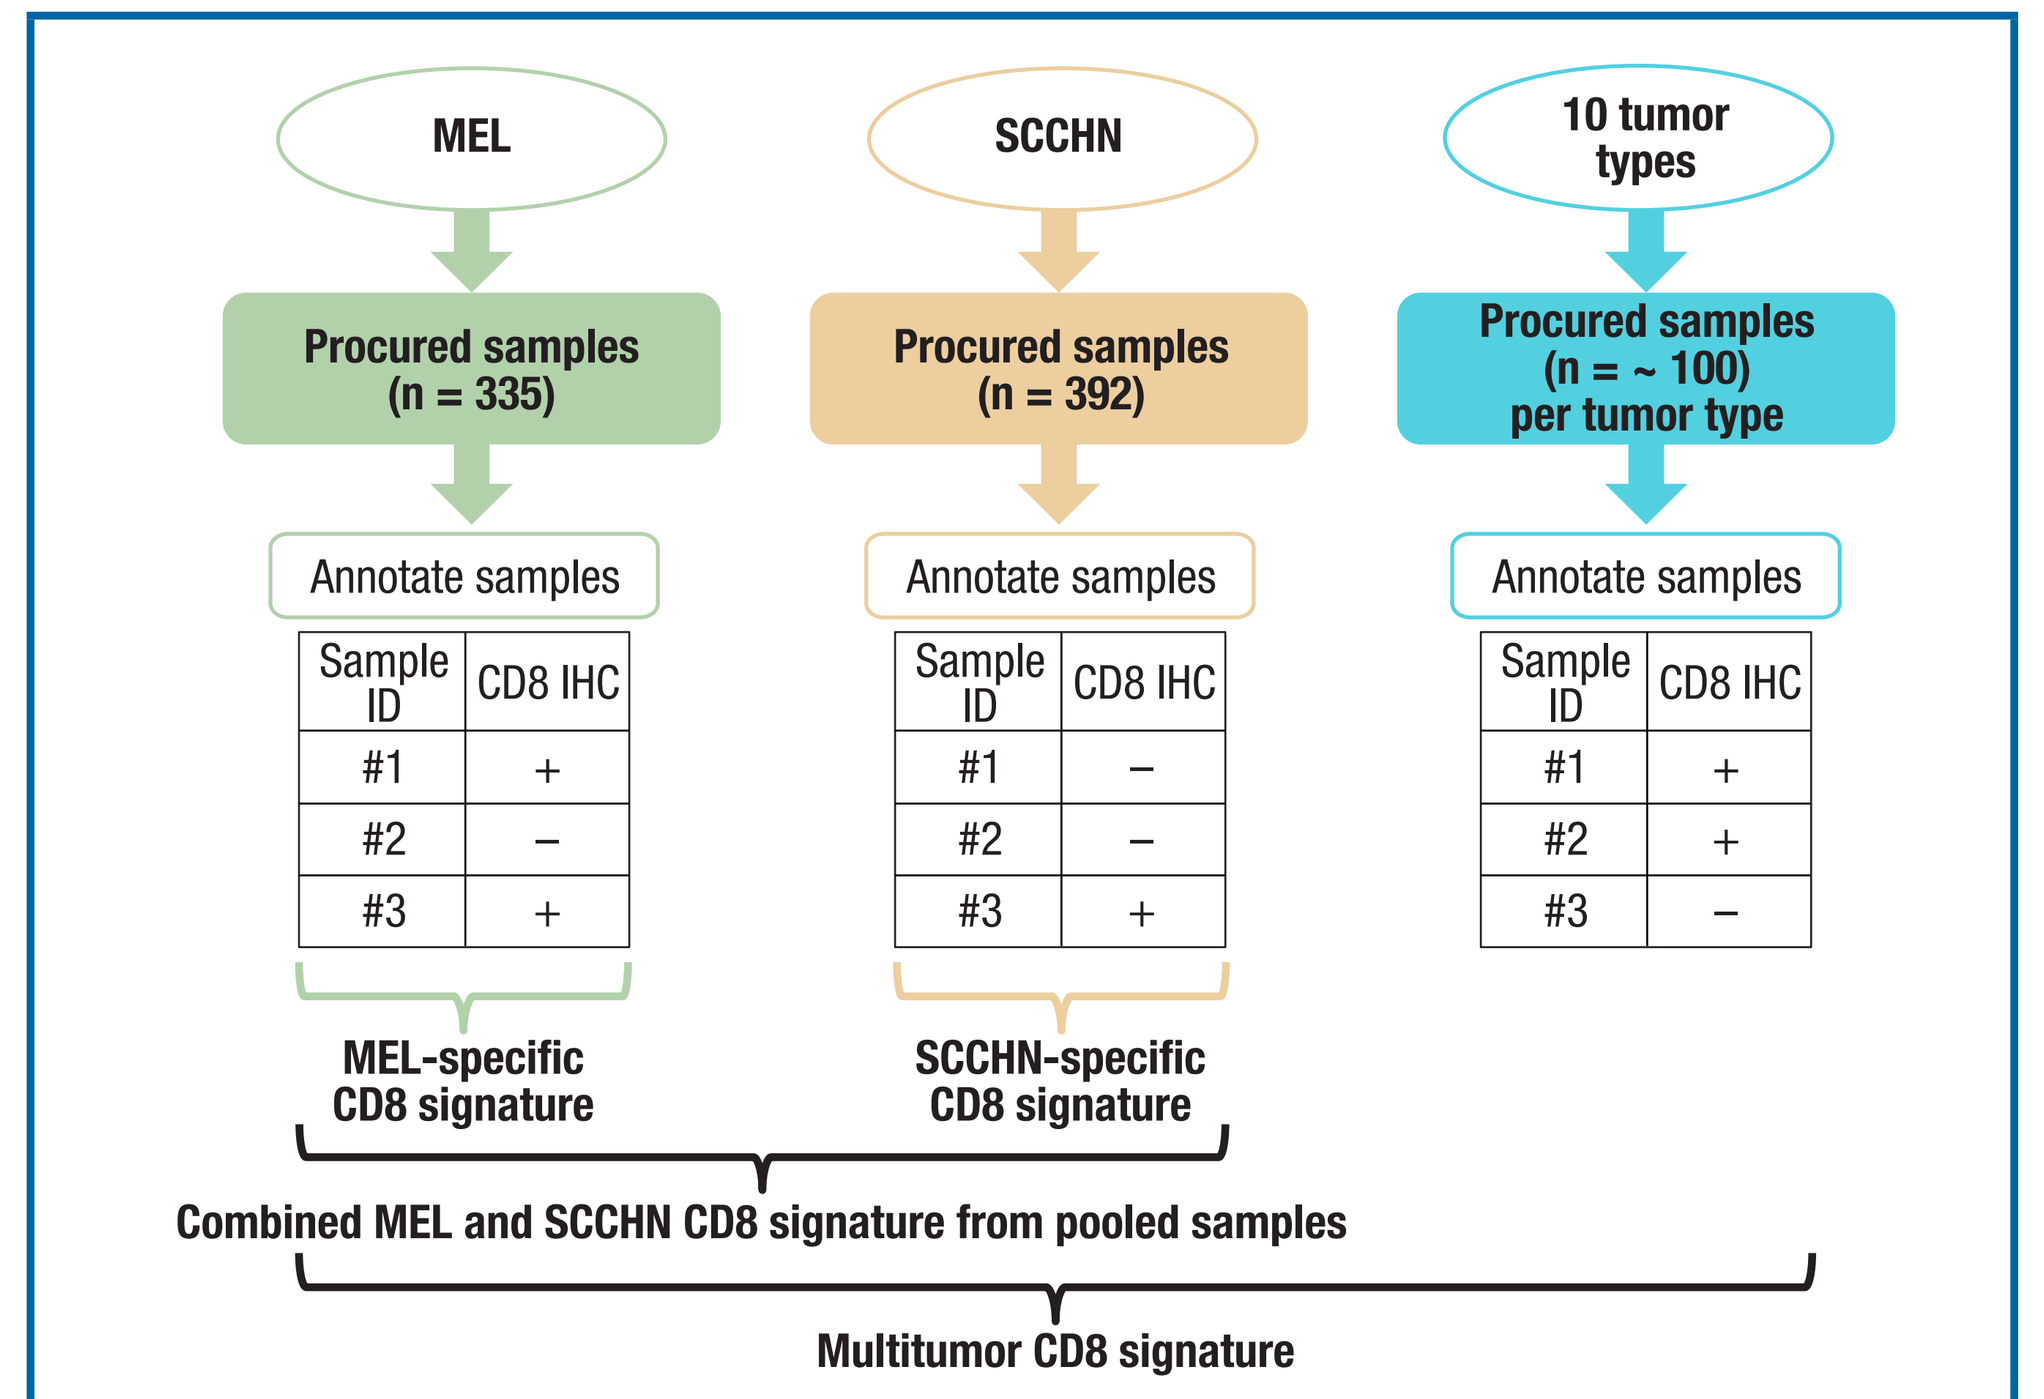
MEL
SCCHN
10 tumor types
Procured samples (n = 335)
Procured samples (n = 392)
Procured samples (n = ~ 100) per tumor type
Annotate samples
Sample ID | CD8 IHC
#1 + / #2 – / #3 +
#1 – / #2 – / #3 +
#1 + / #2 + / #3 –
MEL-specific CD8 signature
SCCHN-specific CD8 signature
Combined MEL and SCCHN CD8 signature from pooled samples
Multitumor CD8 signature

The + and – symbols indicate the range of CD8 positivity, based on scoring of each sample in the dataset by digital pathology. Sample ID, sample identity.

Figure 2. CD8 expression by IHC and CD8 signature score across tumor types

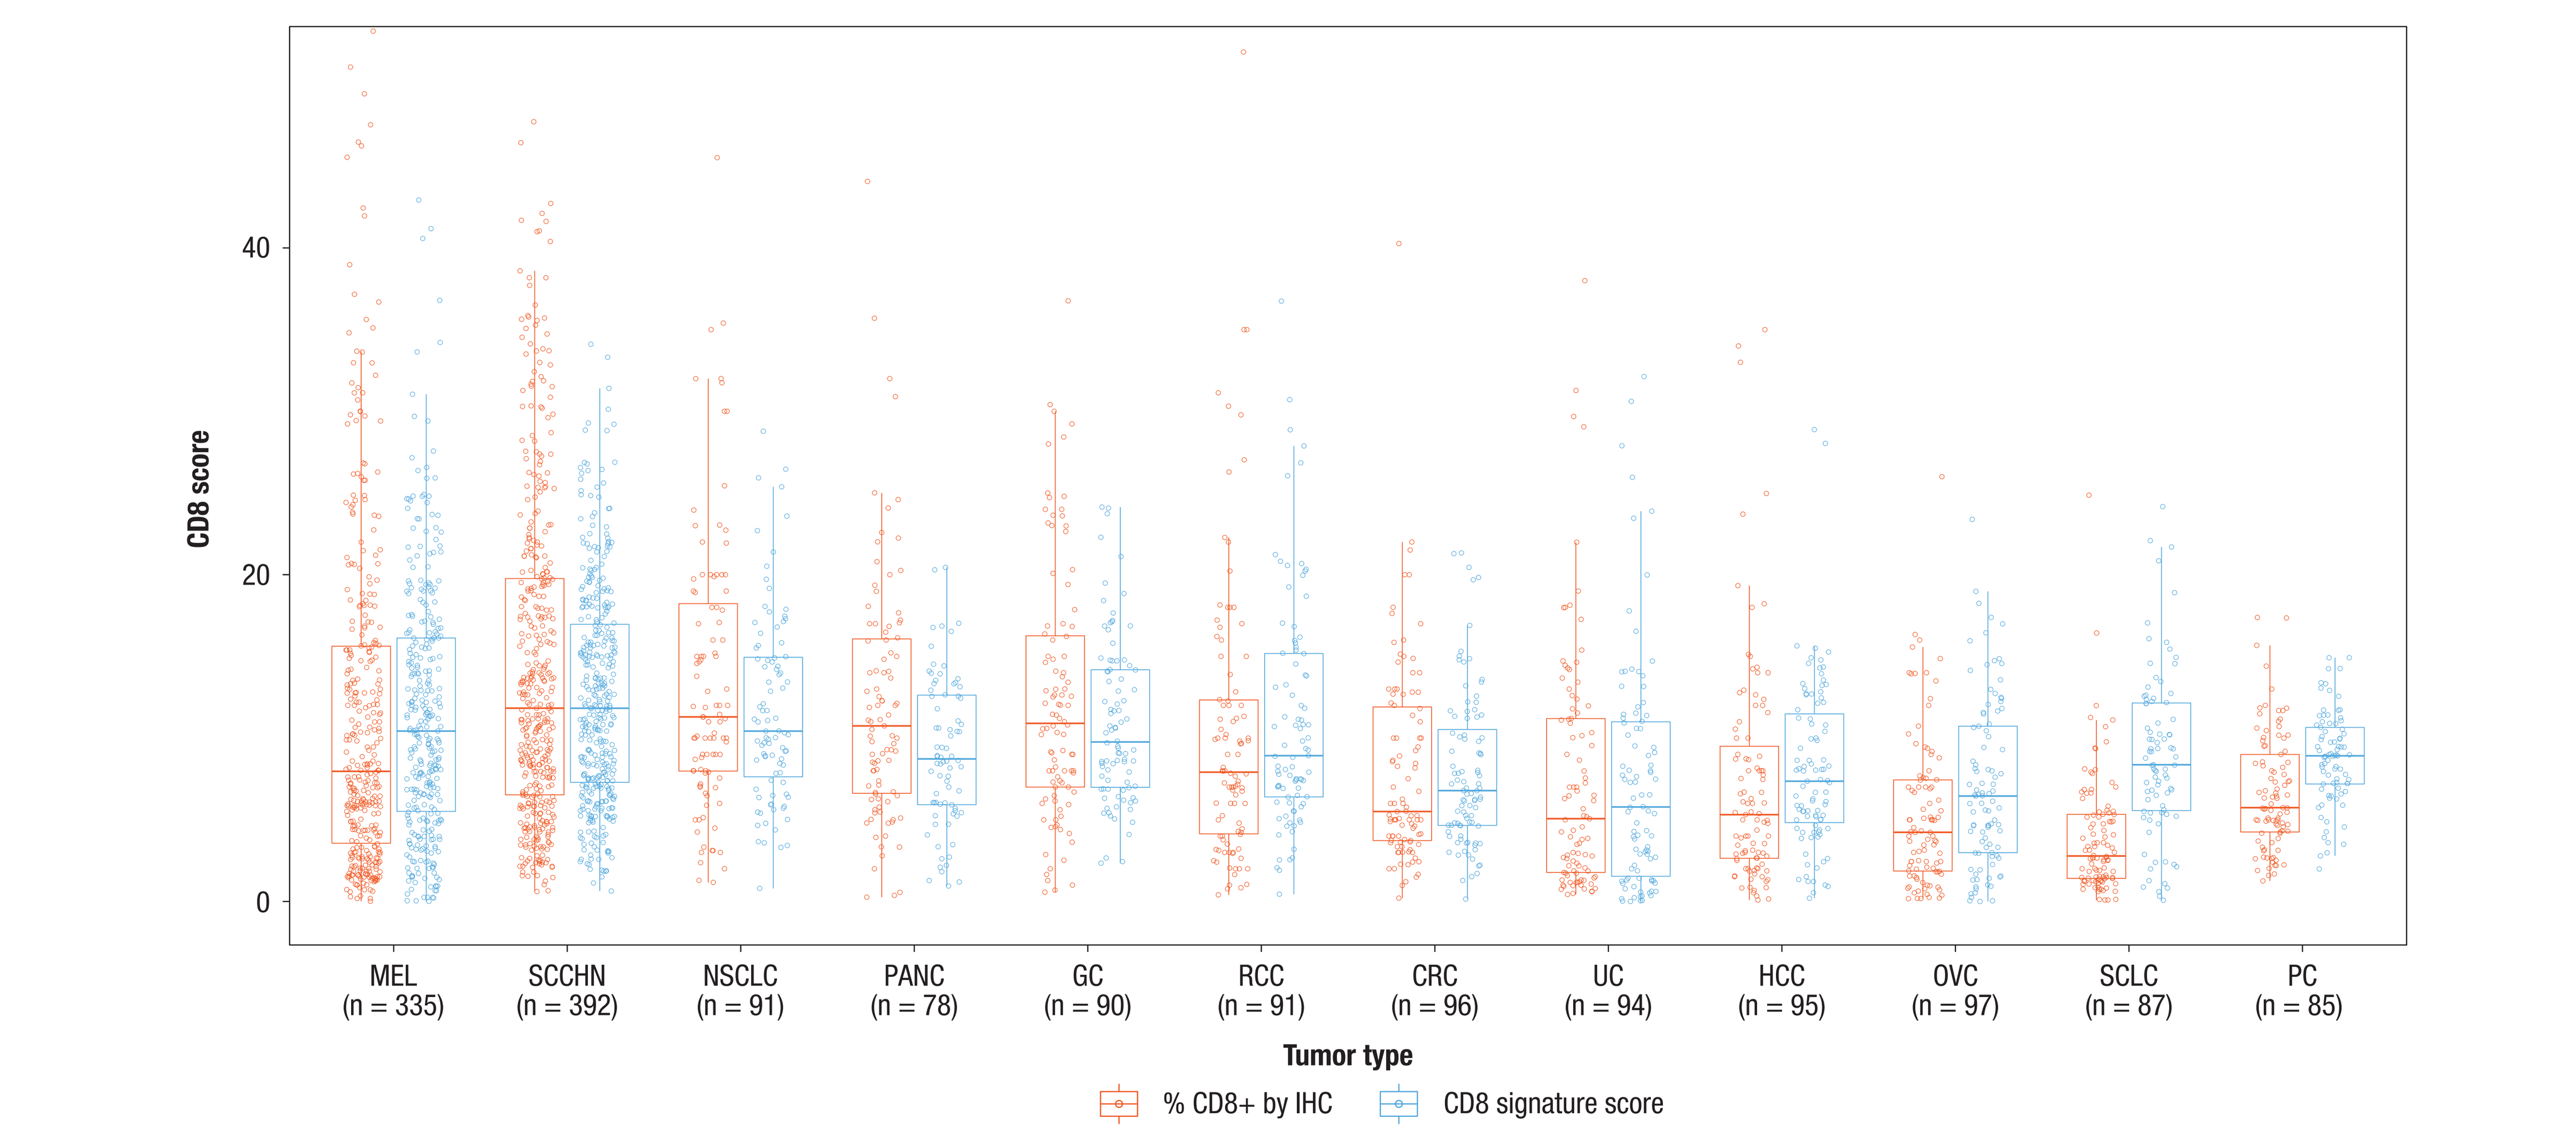
CD8 score
0
20
40
MEL (n = 335)
SCCHN (n = 392)
NSCLC (n = 91)
PANC (n = 78)
GC (n = 90)
RCC (n = 91)
CRC (n = 96)
UC (n = 94)
HCC (n = 95)
OVC (n = 97)
SCLC (n = 87)
PC (n = 85)
Tumor type
% CD8+ by IHC
CD8 signature score

CD8 score represents the CD8 IHC or CD8 signature scores. Each box represents the 25%–75% interquartile range (IQR) and the line is the median value. Whiskers represent values 1.5 × the upper and lower limits of the IQR. CRC, colorectal cancer; GC, gastric cancer; HCC, hepatocellular carcinoma; MEL, melanoma; NSCLC, non-small cell lung cancer; OVC, ovarian cancer; PANC, pancreatic cancer; PC, prostate cancer; RCC, renal cell carcinoma; SCCHN, squamous cell carcinoma of the head and neck; SCLC, small cell lung cancer; UC, urothelial carcinoma.

Results

- Range of CD8 expression, via IHC and the CD8 signature score, was consistent across most tumor types (**Figure 2**)
 - Lower inflammation range and scores were observed in tumor types with lower CD8 expression by IHC (small cell lung cancer [SCLC], ovarian cancer [OVC], and prostate cancer [PC])
 - Higher inflammation range and scores were observed in SCCHN, MEL, non-small cell lung cancer (NSCLC), and gastric cancer (GC)
- Inflammation based on the CD8 signature scores correlated with CD8 IHC scores across multiple tumor types (**Figure 3**)
 - $R^2 \times 100$ values reflecting % variation in CD8 IHC associated with CD8 signature score ranged from 62.41% to 79.21% ($P < 0.0001$) for all tumor types except PC (32.49%)
 - The modest linear correlation of CD8 IHC and CD8 signature score observed in PC samples was consistent with low prevalence of inflammation by CD8 IHC
- The CD8 signature accurately predicted CD8 IHC across tumor types
 - ~ 95% of CD8 IHC/CD8 signature scores fell within the 95% CI derived from the MEL/SCCHN model, except for SCLC (**Figure 3**)
 - For SCLC, 77% of CD8 IHC/CD8 signature scores fell within the 95% CI
- Prevalence of inflammation, by CD8 IHC and the CD8 signature score, was similar in most tumor types except PC and SCLC (**Figure 4**)
 - Prevalence of CD8 score defined by CD8 IHC or CD8 signature score is a function of the cutoff used
- CD8 signature scores were assessed in 15 replicates of 8 samples representative of the dynamic range of inflammation in each of the 12 tumor types
 - Combined precision values across all tumor types are shown as a function of increasing CD8 signature score (**Figure 5**)

Figure 3. Correlation of CD8 IHC and CD8 signature scores across tumor types

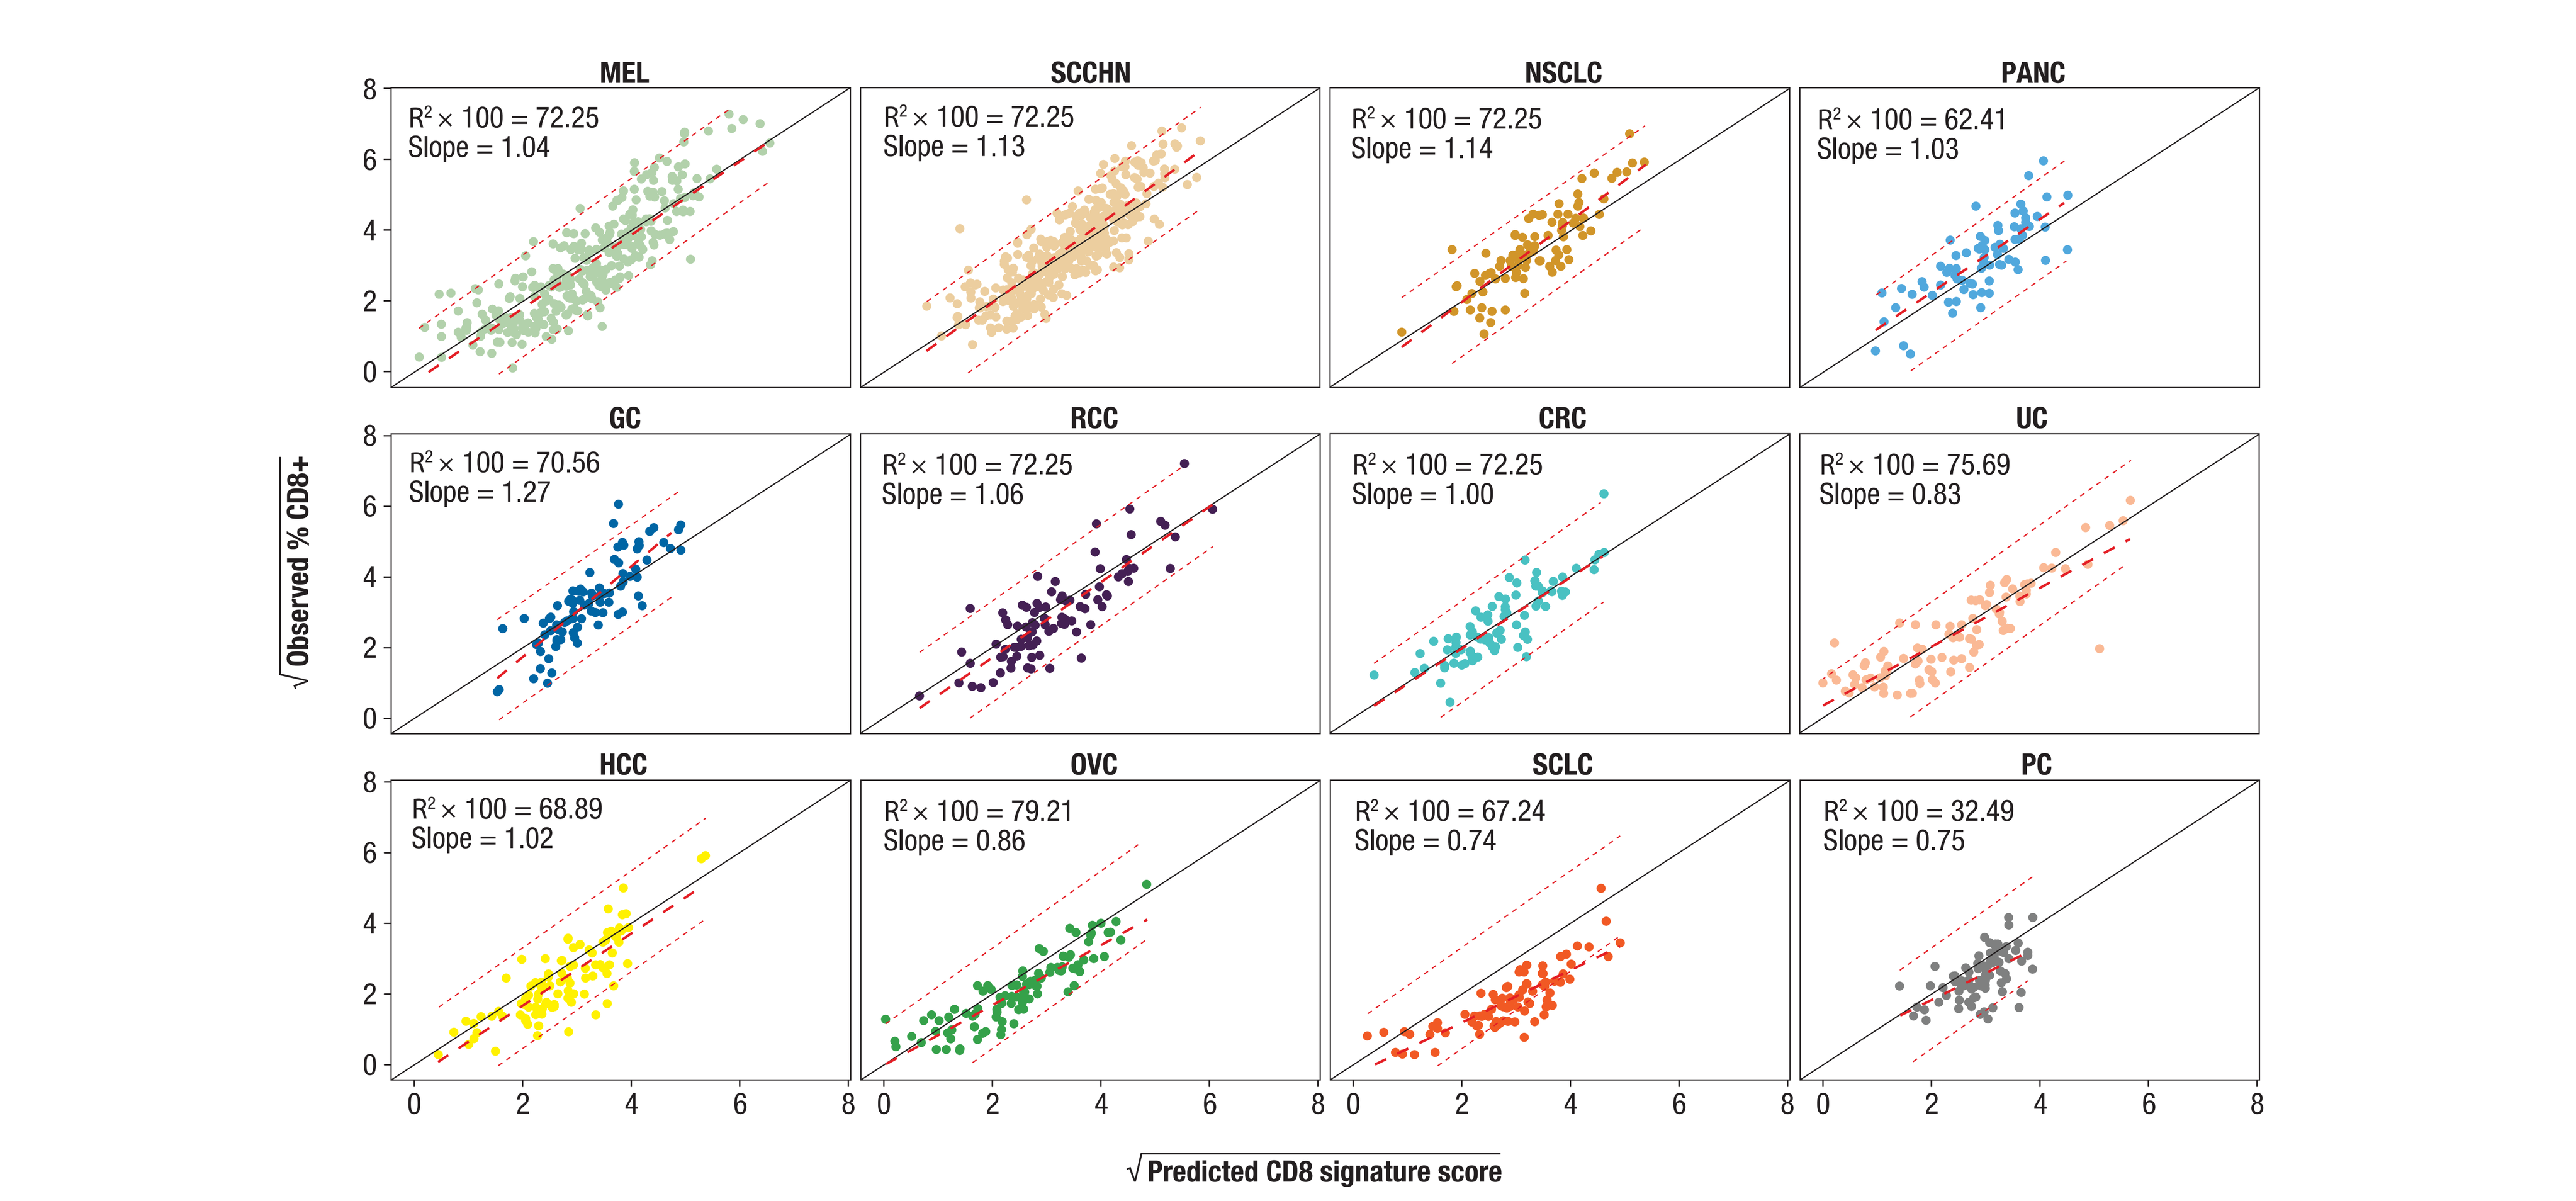
√Observed % CD8+
√Predicted CD8 signature score
MEL $R^2 \times 100 = 72.25$ Slope = 1.04
SCCHN $R^2 \times 100 = 72.25$ Slope = 1.13
NSCLC $R^2 \times 100 = 72.25$ Slope = 1.14
PANC $R^2 \times 100 = 62.41$ Slope = 1.03
GC $R^2 \times 100 = 70.56$ Slope = 1.27
RCC $R^2 \times 100 = 72.25$ Slope = 1.06
CRC $R^2 \times 100 = 72.25$ Slope = 1.00
UC $R^2 \times 100 = 75.69$ Slope = 0.83
HCC $R^2 \times 100 = 68.89$ Slope = 1.02
OVC $R^2 \times 100 = 79.21$ Slope = 0.86
SCLC $R^2 \times 100 = 67.24$ Slope = 0.74
PC $R^2 \times 100 = 32.49$ Slope = 0.75

CRC, colorectal cancer; GC, gastric cancer; HCC, hepatocellular carcinoma; MEL, melanoma; NSCLC, non-small cell lung cancer; OVC, ovarian cancer; PANC, pancreatic cancer; PC, prostate cancer; RCC, renal cell carcinoma; SCCHN, squamous cell carcinoma of the head and neck; SCLC, small cell lung cancer; UC, urothelial carcinoma.

Figure 4. Prevalence of inflammation by CD8 IHC and CD8 signature scores across tumor types

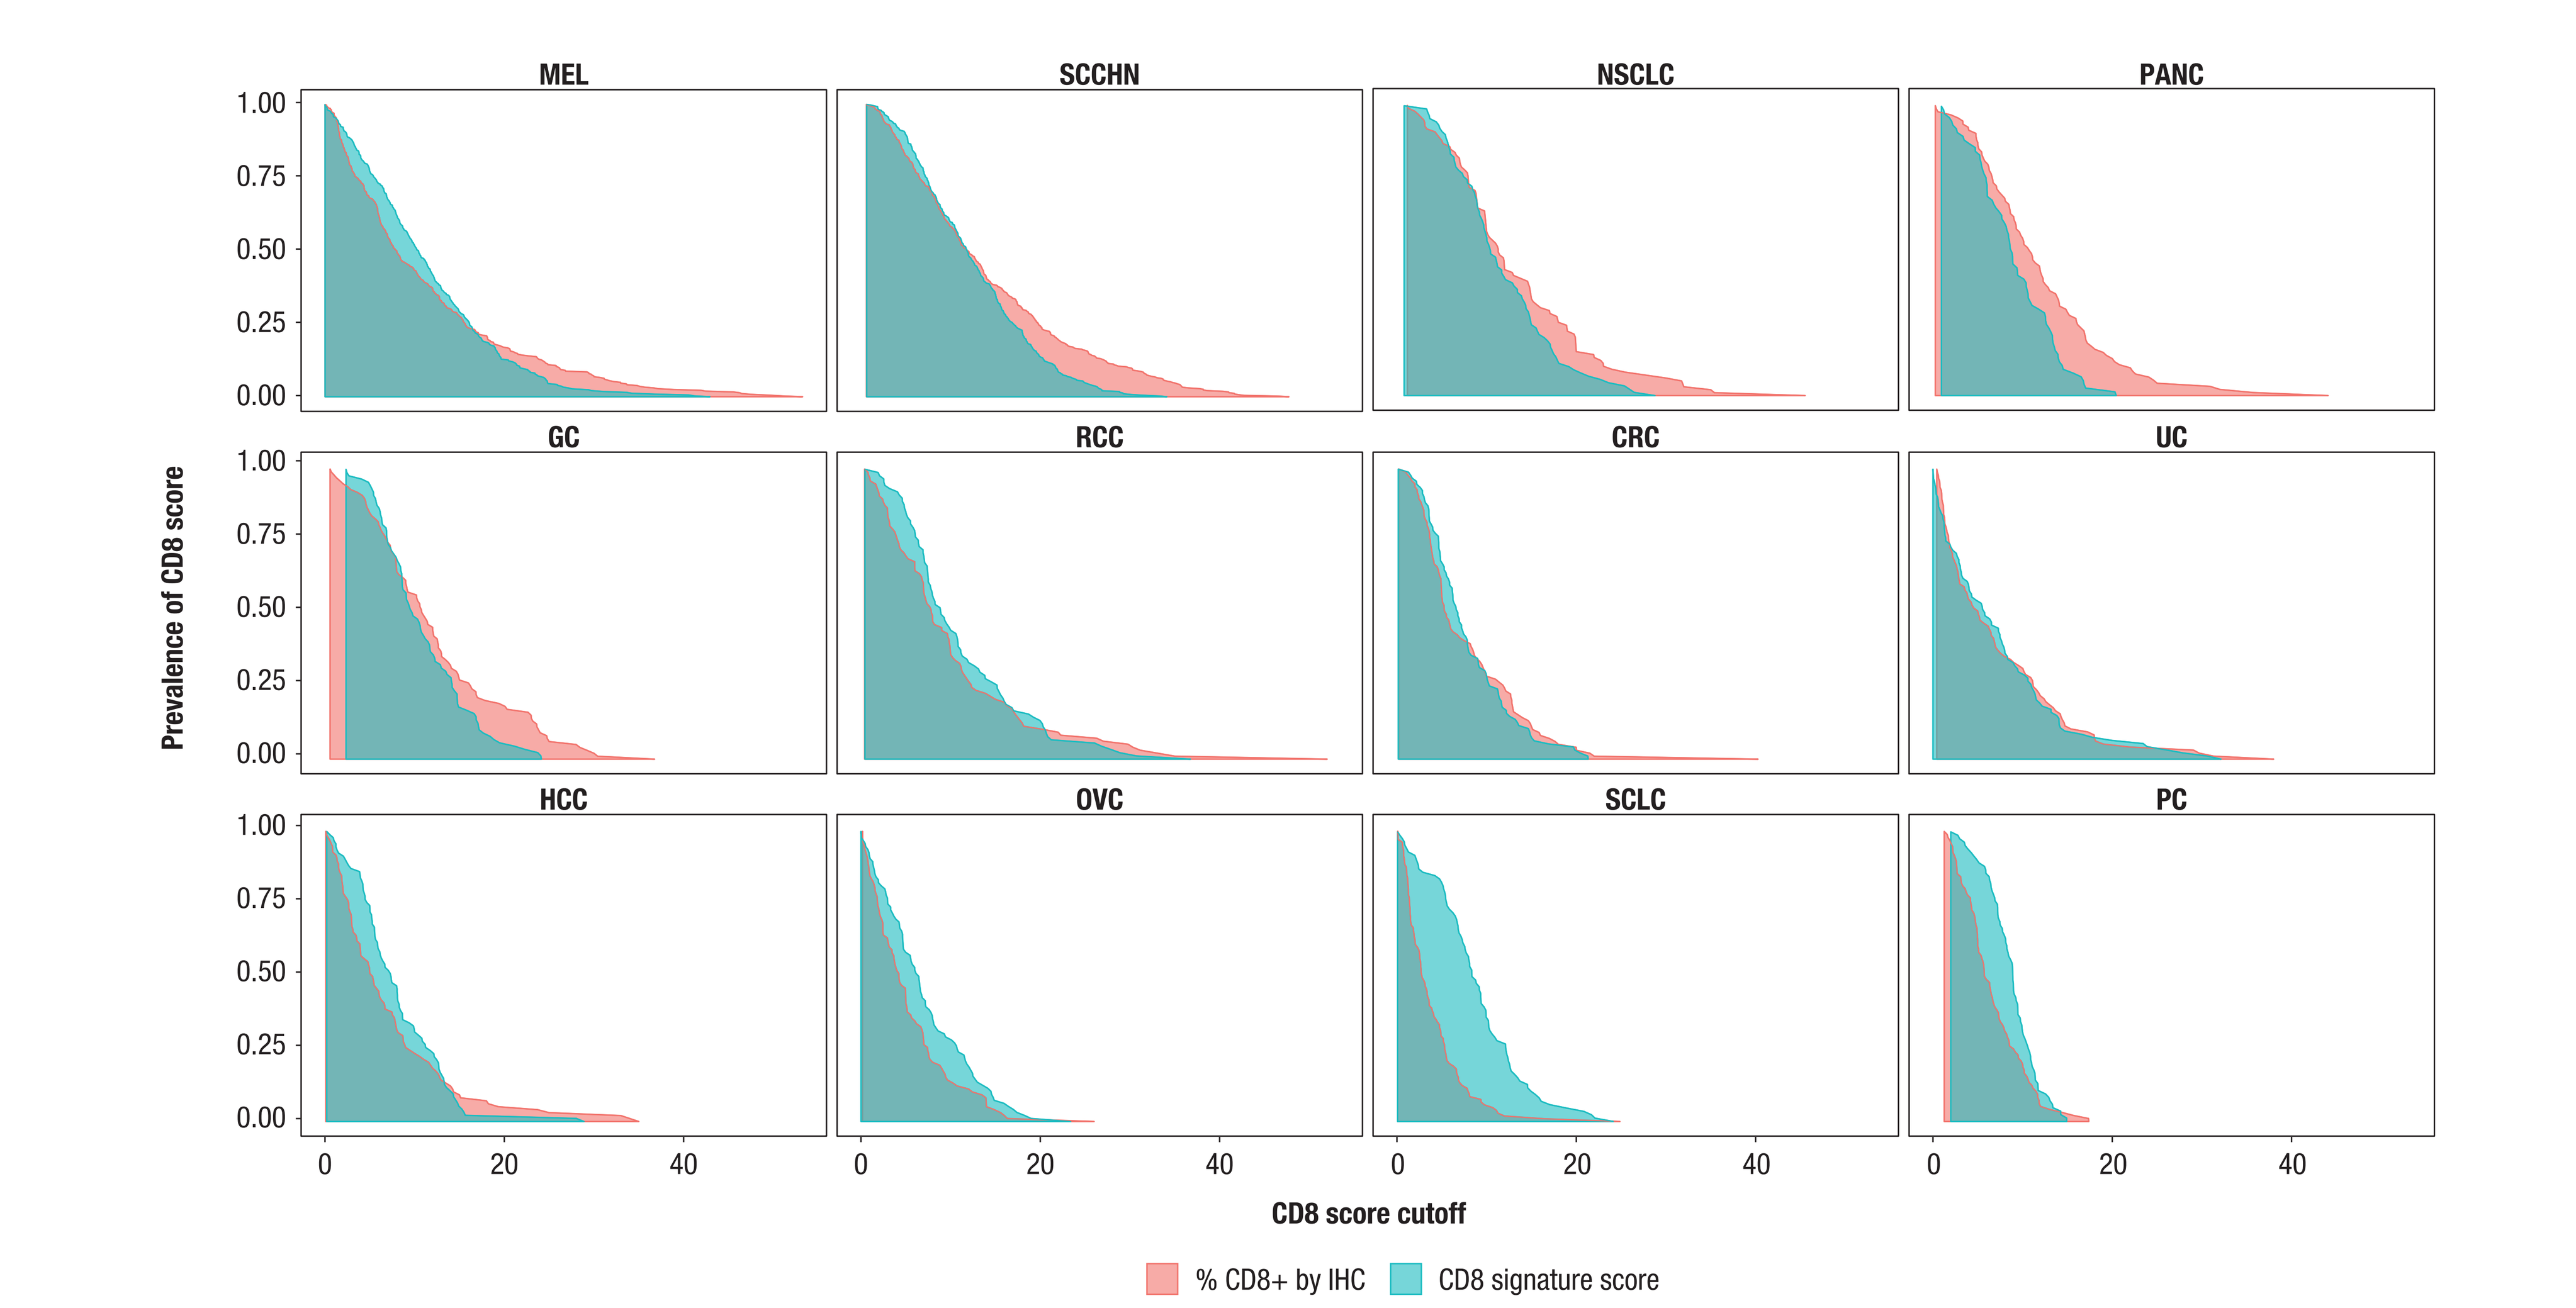
Prevalence of CD8 score
CD8 score cutoff
MEL
SCCHN
NSCLC
PANC
GC
RCC
CRC
UC
HCC
OVC
SCLC
PC
% CD8+ by IHC
CD8 signature score

CD8 score represents the CD8 IHC or CD8 signature scores. CRC, colorectal cancer; GC, gastric cancer; HCC, hepatocellular carcinoma; MEL, melanoma; NSCLC, non-small cell lung cancer; OVC, ovarian cancer; PANC, pancreatic cancer; PC, prostate cancer; RCC, renal cell carcinoma; SCCHN, squamous cell carcinoma of the head and neck; SCLC, small cell lung cancer; UC, urothelial carcinoma.

Figure 5. Precision of the CD8 signature improves with increasing score across multiple tumor types

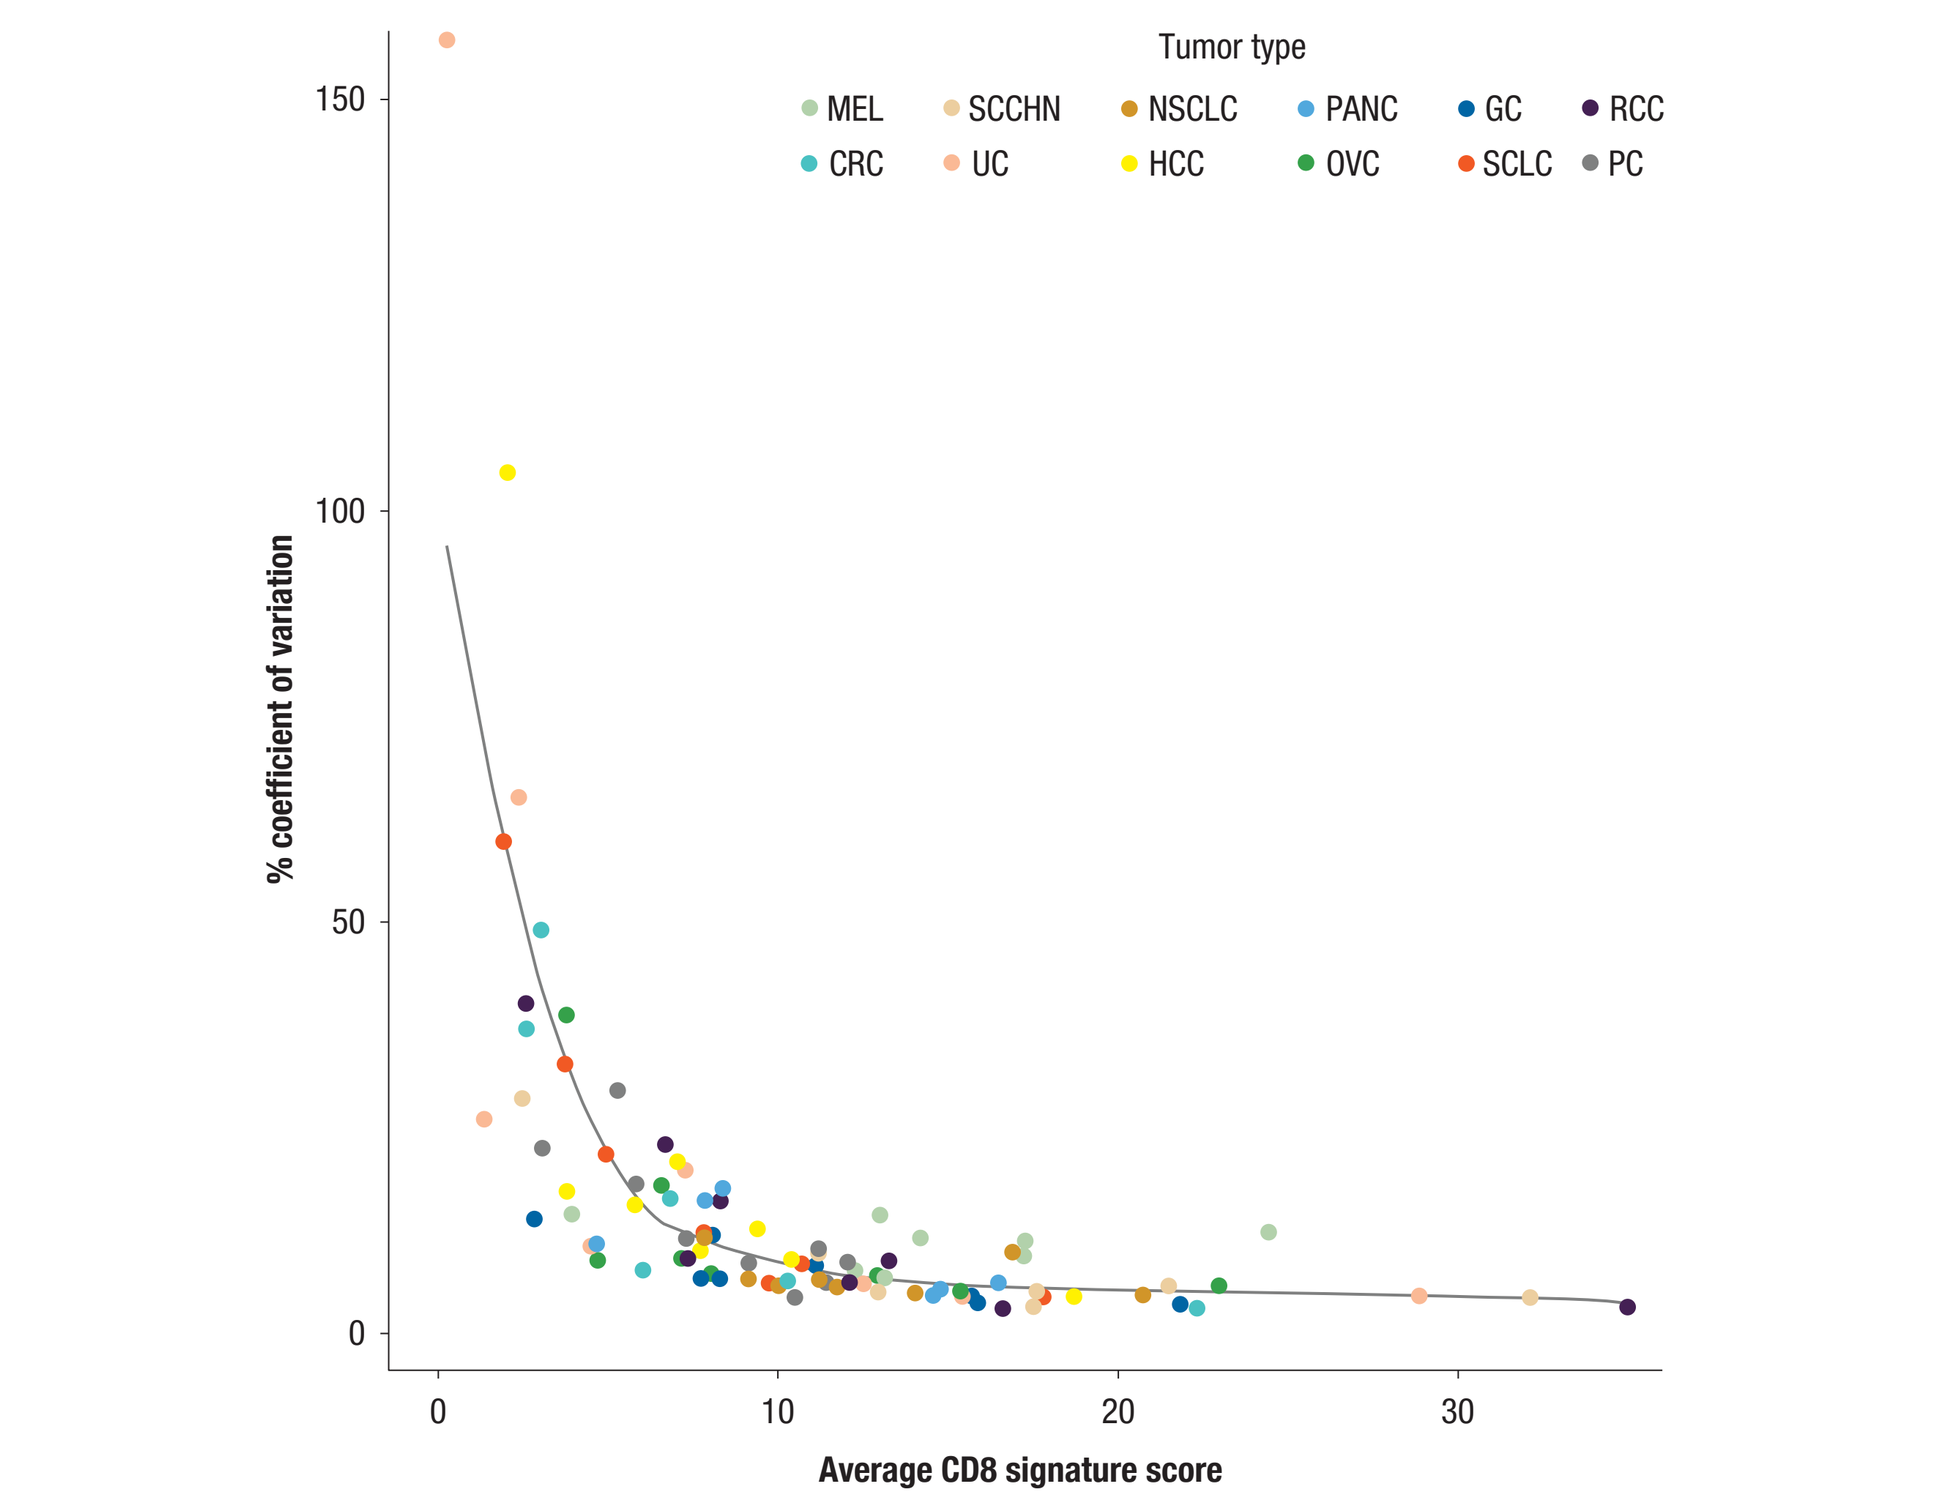
Tumor type
MEL
SCCHN
NSCLC
PANC
GC
RCC
CRC
UC
HCC
OVC
SCLC
PC
% coefficient of variation
Average CD8 signature score

Precision of CD8 signature scores was assessed using 8 samples with 15 replicates per tumor type. CRC, colorectal cancer; GC, gastric cancer; HCC, hepatocellular carcinoma; MEL, melanoma; NSCLC, non-small cell lung cancer; OVC, ovarian cancer; PANC, pancreatic cancer; PC, prostate cancer; RCC, renal cell carcinoma; SCCHN, squamous cell carcinoma of the head and neck; SCLC, small cell lung cancer; UC, urothelial carcinoma.

Conclusions

- We have developed a GEP-based, IUO inflammation assay with the potential to be utilized prospectively in a clinical trial setting
- Using this 95-gene panel assay, we have derived a 16-gene CD8 signature that can accurately assess inflammation in the TME and has been validated across 12 tumor types
 - Inflammation status determined by the CD8 signature score correlated with that derived from analyzing CD8 expression by IHC in all tumor types tested except PC. This may be a result of a smaller range of CD8 IHC and CD8 signature scores in PC samples and warrants further investigation
- Prevalence of inflammation characterized by CD8+ T cells in the TME across multiple tumor types, using IHC and a CD8 signature using GEP, was similar for most tumor types, indicating that this gene signature may be utilized to accurately quantify CD8 expression

References

1. Darvin P, et al. *Exp Mol Med* 2018;50(12). doi: 10.1038/s12276-018-0191-1.
2. Stoll G, et al. *Oncotarget* 2015;6:11894–11909.
3. Barnes TA, et al. *Br J Cancer* 2017;117:451–460.
4. Sharma P, et al. *Lancet Oncol* 2017;18:312–322.
5. Cristescu R, et al. *Science* 2018;362:eaar3593.

Acknowledgments

- The study was supported by Bristol-Myers Squibb, HTG Molecular Diagnostics Inc, and QIAGEN
- The authors would like to acknowledge the contributions of Julie Carman, Keyur Desai, Saumya Pant, Gaurav Shah, Kim Zerba, and Nancy Zhang from Bristol-Myers Squibb; Alistair English, Michael Evershed, and Pendeep Gill from QIAGEN; and Nageswara Rao Alla, David Kroeppler, Steven Lung, Di Ran, and Michael Reubens from HTG Molecular Diagnostics Inc
- All authors contributed to and approved the presentation; writing and editorial assistance was provided by Katerina Pipili, PhD, and Jay Rathi, MA, of Spark Medica Inc, funded by Bristol-Myers Squibb

Scientific Content On-demand

To request a copy of this poster:

Text "GEPCD8" to +1-404-800-0746

OR

Scan QR code via a barcode reader application

By requesting this content, you agree to receive a one-time communication using automated technology. Msg. & data rates may apply. Links are valid for 30 days after the congress presentation date.

Presented at the American Society of Clinical Oncology Annual Meeting, May 31–June 4, 2019, Chicago, IL

Email: Peter.Szabo@bms.com

Copies of this poster obtained through Quick Response (QR) Code or text message are for personal use only and may not be reproduced without written permission from ASCO® and the author of this poster.